Clinical Trial Simulation to Estimate the Sample Size for Investigation of the Impact of Drug A on the Pharmacokinetics of Methotrexate, commonly co-administered in Rheumatoid Arthritis (RA)

Shuying Yang and Misba Beerahee
Clinical Pharmacology Modelling and Simulation, GSK, UK

Introduction

Methotrexate (MTX) is widely used as first line DMARD in RA patients. It was observed that frequent use of MTX can cause serious toxicities, even though no conclusive correlation with MTX drug levels is reported (Bannwarth et al. 1996). Drug A has a novel mechanism of action for potential treatment in RA and is at the planning stage of a large dose range phase IIb trial. Although a risk-assessment analysis showed little, if any, metabolic liability for interaction between Drug A and MTX, there is, nevertheless, a regulatory as well as commercial need to capture or discharge any potential risk for PK drug-drug interaction (DDI). Instead of running a standard DDI study before PIIb, we propose to embed a sub-study within the planned phase IIb trial, obtaining sparse MTX PK samples to address any impact of drug A on the systemic exposure of MTX.
The aim of this simulation work was to assess the power of a parallel group bioequivalence sub-study within the phase IIb trial, using population PK modelling approach with sparse PK samples.

Methods

Figure 1 shows the methods and strategies for this clinical trial simulation. The following five steps are involved in the simulation. The simulations and estimations are conducted using WFN (Holford) linking with NONMEM VI (Beal et. al 1989-2006).

Step 1) Derive a population PK model for MTX;
Step 2) Determine the optimal sampling times under model determined in step 1;
Step 3) Simulate individual PK data based on the model and sampling times for different scenarios;
Step 4) Estimate PK parameters from each simulated data set;
Step 5) Summarise the estimated parameters

Population PK Model of MTX

Population PK model of MTX in RA population was developed using non-linear mixed effect method based on in-house study with patients on stable MTX (10-25mg weekly). Total of 15 subjects with 12 to 20 blood samples per subject were available for model building. The final model was a 2-compartment 1st order absorption and elimination with proportional error. The model parameterisation is shown in Figure 1 and the estimates are presented in the Table below

Parameters	THETA (95%CI)	Ω(SE)
CL (L/h)	11.59 (10.28, 13.07)	0.0515 (0.0175)
Vc (L)	36.97 (32.79, 41.68)	0.0539 (0.0165)
Vp (L)	9.12 (7.61, 10.91)	-
Q (L/h)	1.20 (0.97, 1.48)	-
Ka (1/h)	4.31 (2.97, 6.23)	0.47 (0.212)
SIGMA*	0.042 (0.00867)	

* Estimate (SE)

Sampling Times

Initially, optimal sparse sampling times from the population PK model for Drug A were estimated using the optimal design software PopDes (Gueorguieva 2007). Slight modifications to the sampling times were needed for flexibility within the clinical trial setting. The modified sampling times were 5min, 1, 6, 12, and 24 h post dose. It was estimated that over 90% efficiency would be achieved with this sampling scheme.

In the simulation, flexible sampling windows within about 2 h of these modified optimal sampling times were allowed to mimic the real trial situations.

Figure 1: Methods and strategies for the simulation study

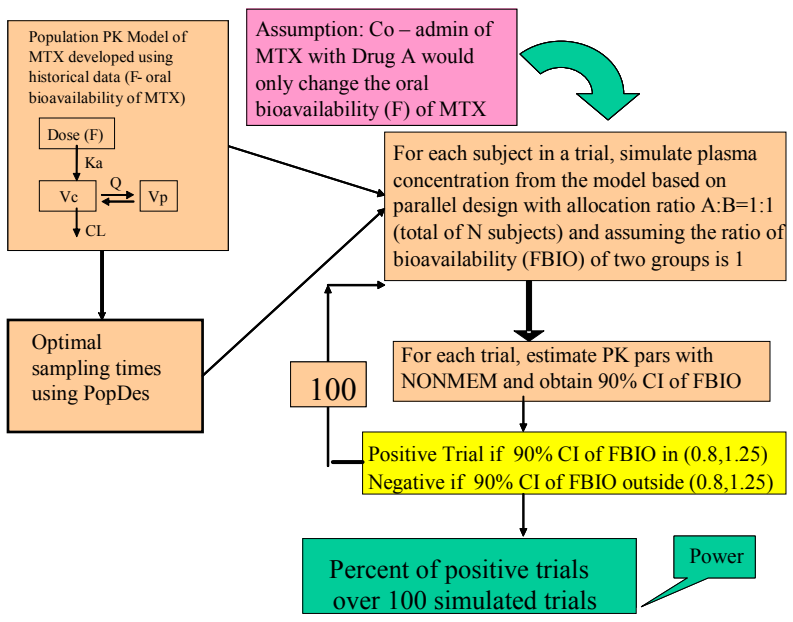
Population PK Model of MTX developed using historical data (F- oral bioavailability of MTX)
Dose (F)
Ka
Vc
Q
Vp
CL
Assumption: Co – admin of MTX with Drug A would only change the oral bioavailability (F) of MTX
For each subject in a trial, simulate plasma concentration from the model based on parallel design with allocation ratio A:B=1:1 (total of N subjects) and assuming the ratio of bioavailability (FBIO) of two groups is 1
Optimal sampling times using PopDes
100
For each trial, estimate PK pars with NONMEM and obtain 90% CI of FBIO
Positive Trial if 90% CI of FBIO in (0.8,1.25)
Negative if 90% CI of FBIO outside (0.8,1.25)
Percent of positive trials over 100 simulated trials
Power

Study Design of MTX DDI Sub-study

The embedded MTX DDI sub-study in the PIIb is designed as a double-blind, randomised parallel group in RA patients. The double-blind ensures the integrity of the PIIb study, with a stratified randomisation based on subject treatment with MTX at entry in the PIIb study.
The MTX DDI is focussed on the group of subjects who are stabilised on MTX. The schematic of the design is shown in Figure 2. After screening, subjects are randomised with equal allocation ratio, into each of the active treatment group or placebo group, stratified by their treatment on MTX.

Figure 2: Schematic View of the Proposed Study Design of the sub-study

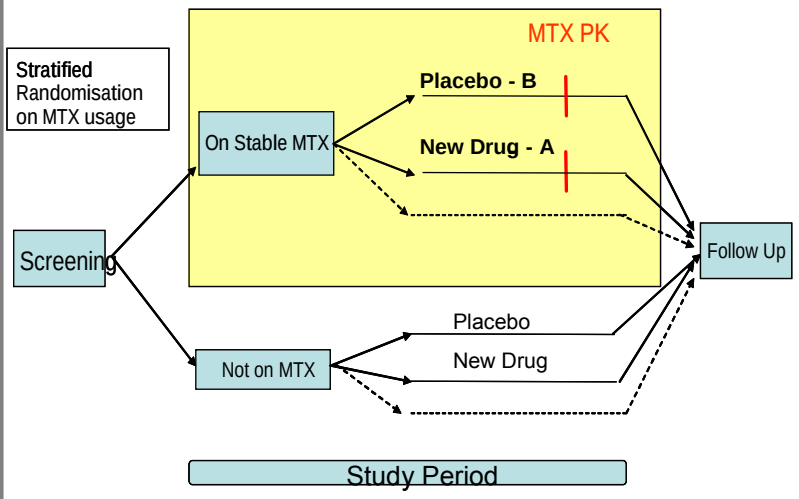
Stratified Randomisation on MTX usage
MTX PK
Placebo - B
New Drug - A
On Stable MTX
Screening
Follow Up
Placebo
New Drug
Not on MTX
Study Period

Hypothesis

Preliminary investigation of metabolic pathways indicated that any likely effect (if at all) of drug A on MTX PK would be on the oral bioavailability.
Let FBIO indicate the relative bioavailability of the two groups, A: TMT=1, B: TMT=0 and F1=(1-TMT)+TMT*FBIO.
The hypothesis for testing the equivalence is:

- H0: FBIO<=RL or FBIO>=RU, not equivalent
- H1: RL<FBIO<RU, equivalent
- (RL,RU)= (0.8, 1.25)

Simulations

Four scenarios were investigated:

- **Low IIV Low Res (I)**: Ω_{CL} =0.0515 Ω_{Vc} = 0.0539, SIGMA=0.042 (from in house data)
- **High IIV High Res (II)**: Ω_{CL} =0.14 Ω_{Vc} = 0.08, SIGMA=0.076 (from literature Godfrey 1998)
- **Low IIV High Res (III)**: Ω_{CL} =0.0515 Ω_{Vc} =0.0539, SIGMA=0.076
- **High IIV Low Res (IV)**: Ω_{CL} =0.14 Ω_{Vc} = 0.08, SIGMA=0.042

For each scenario, N=40, 60, 70, and 80 subjects in a trial were evaluated. Total of 100 trials were simulated for each scenario and N.

Results

Power Assessment

- Positive trial (equivalence holds) - the 90% CI of FBIO within (0.8,1.25).
- For each scenario and each N, the power of the design is calculated as the percent of positive results over the 100 simulated trials. (Figure 3)

Figure 3: Estimated Power by total number of subjects with different scenarios

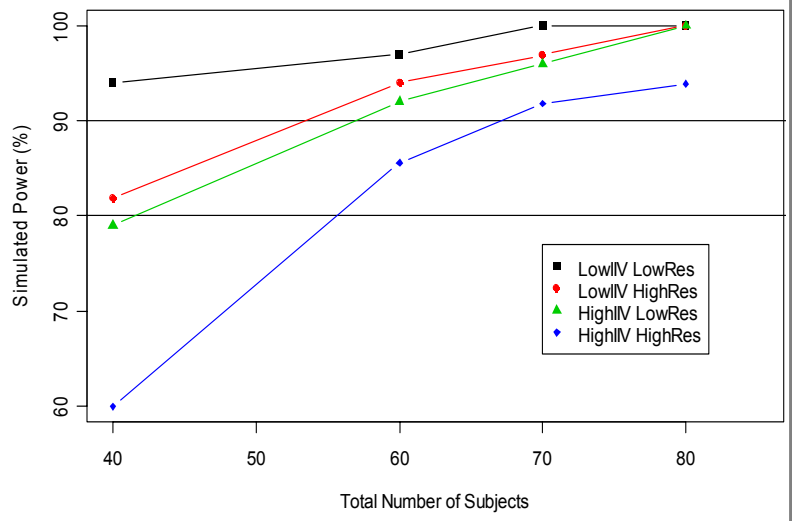
Simulated Power (%)
100
90
80
70
60
40
50
60
70
80
Total Number of Subjects
LowIIV LowRes
LowIIV HighRes
HighIIV LowRes
HighIIV HighRes

Bias Assessment

- The bias of the population PK modelling with sparse samples was assessed
- Figure 4 shows a summary of the percent bias calculated by: Bias%=((estimates – true value)/true value)*100

Figure 4: Percent bias of PK parameters Scenario II with N=70.

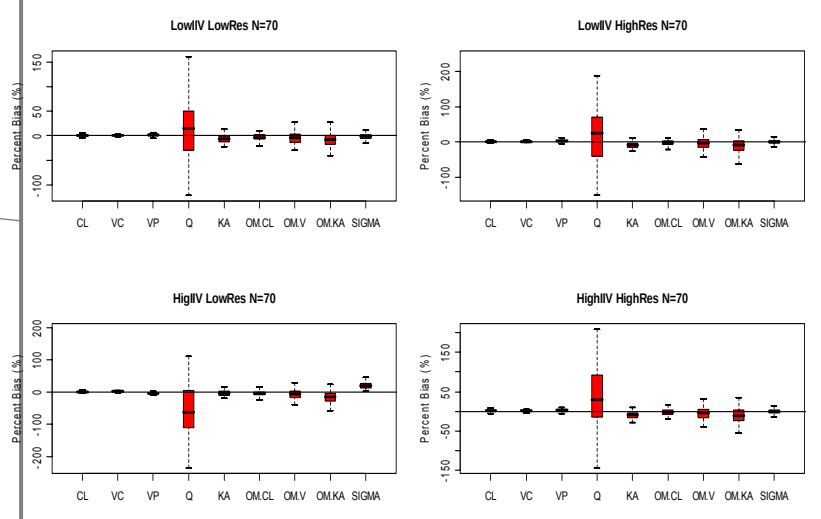
LowIIV LowRes N=70
LowIIV HighRes N=70
HigIIV LowRes N=70
HighIIV HighRes N=70
Percent Bias (%)
CL
VC
VP
Q
KA
OM.CL
OM.V
OM.KA
SIGMA

Summary

- Prospective evaluation of design of sub-study, such as DDI, within a large patient study can be invaluable from ethical and streamlined efficient drug development perspective.
- A total of 70 subjects would provide over 90% power to test for drug-drug interaction with MTX based on the worst case scenario (II).
- An adaptive sample size adjustment design could also be implemented following re-assessment of the variability of MTX levels in this sub-study in a blinded manner (e.g based on initial 20 patients).
- The use of population PK modelling with sparse data for addressing PK DDI is appropriate and is more cost-effective than standard NCA approach (requires 120 subjects to reach 90% power)
- Clinical trial simulation is a flexible tool for the evaluation of sample size, power and the biases for complex trial designs.

References

- Bannwarth B, Clin. Pharmacokinet. 1996, 30, 194
- Nick Holford: Wings for NONMEM, http://wfn.sourceforge.net/index.html.
- Beal SL, et.al (eds) (1989–2006) NONMEM users guides. Icon development solutions. Ellicott City
- Gueorguieva I et al. Computer methods for programs in biomedicine, 2007, 86, 51-61
- Godfrey C, et. al. Br J Clin Pharmacol, 1998, 46, 369-376.

Created by